One Year Follow-up on the First Patient Treated with Nula-Cel:

An Autologous CRISPR/Cas9 *HBB* Gene Corrected CD34⁺ Cell Product to Treat Sickle Cell Disease

David Shyr[3], Robert Lowsky[3], Weston Miller[2], Mark Schroeder[4], Tonia J Buchholz[2], Kirstin Dougall[3], Allison Intondi[2], Alexandra Charles[3], Josh Lehrer[2], Ali Bouge[3], Stacy Wolf[3], Blair McDonald[3], Hena Din[3], Gopin Saini[3], Alana Lerner[2], Manal Amoury[2], Sebastien Treuch[2], Glen Chew[2], Brian Silva[2], Haider Mashhedi[2], Vincent Siu[2], Ian Perrone[2], Twaritha Vijay[2], Aayami Jain[2], Kristina Krassovsky[2], Will Matern[2], Premanjali Lahiri[2], Ryan Rodriquez[2], Jason Skowronski[1,2], Arpit Batish[3], Dana Margittai[3], Prachi Wani[3], Timothy Van Horn[3], Claudia Flautero[3], Rosalva Flores[3], Jade Lee[3], Keri Tate[3], Steven Feldman[3], Maria Grazia Roncarolo[1,2,3], John DiPersio[4], Valentin Barsan[1,3], Matthew Porteus[1,2,3]

[1]Kamau Therapeutics, South San Francisco, CA, USA | [2]Graphite Biologics, South San Francisco, CA, USA | [3]Stanford University, Stanford, CA USA | [4]Washington University, St . Louis, MO USA

Introduction

Sickle cell disease (SCD) is an autosomal recessive genetic disease caused by a single point mutation in both copies of the beta-globin gene, resulting in sickle hemoglobin (HgbS) production instead of adult hemoglobin (HgbA). It causes early mortality with lifelong morbidity for patients and remains a global disease with a large unmet medical need.

Previous allogeneic hematopoietic stem cell transplant data have demonstrated a competitive advantage for homozygous (AA) or heterozygous (AS) hemoglobin erythroid progenitors over sickle disease (SS) erythroid progenitors with curative outcomes.

KMAU-001 (nulabeglogene autogedtemcel) is an investigational, gene-edited, autologous hematopoietic stem cell-based therapy in clinical development for SCD that is designed to directly correct the underlying mutation, thereby decreasing HgbS production and restoring HgbA expression.

High-Efficiency HDR-Editing via CRISPR/HiFi Cas9 Precisely Corrects the SCD Mutation

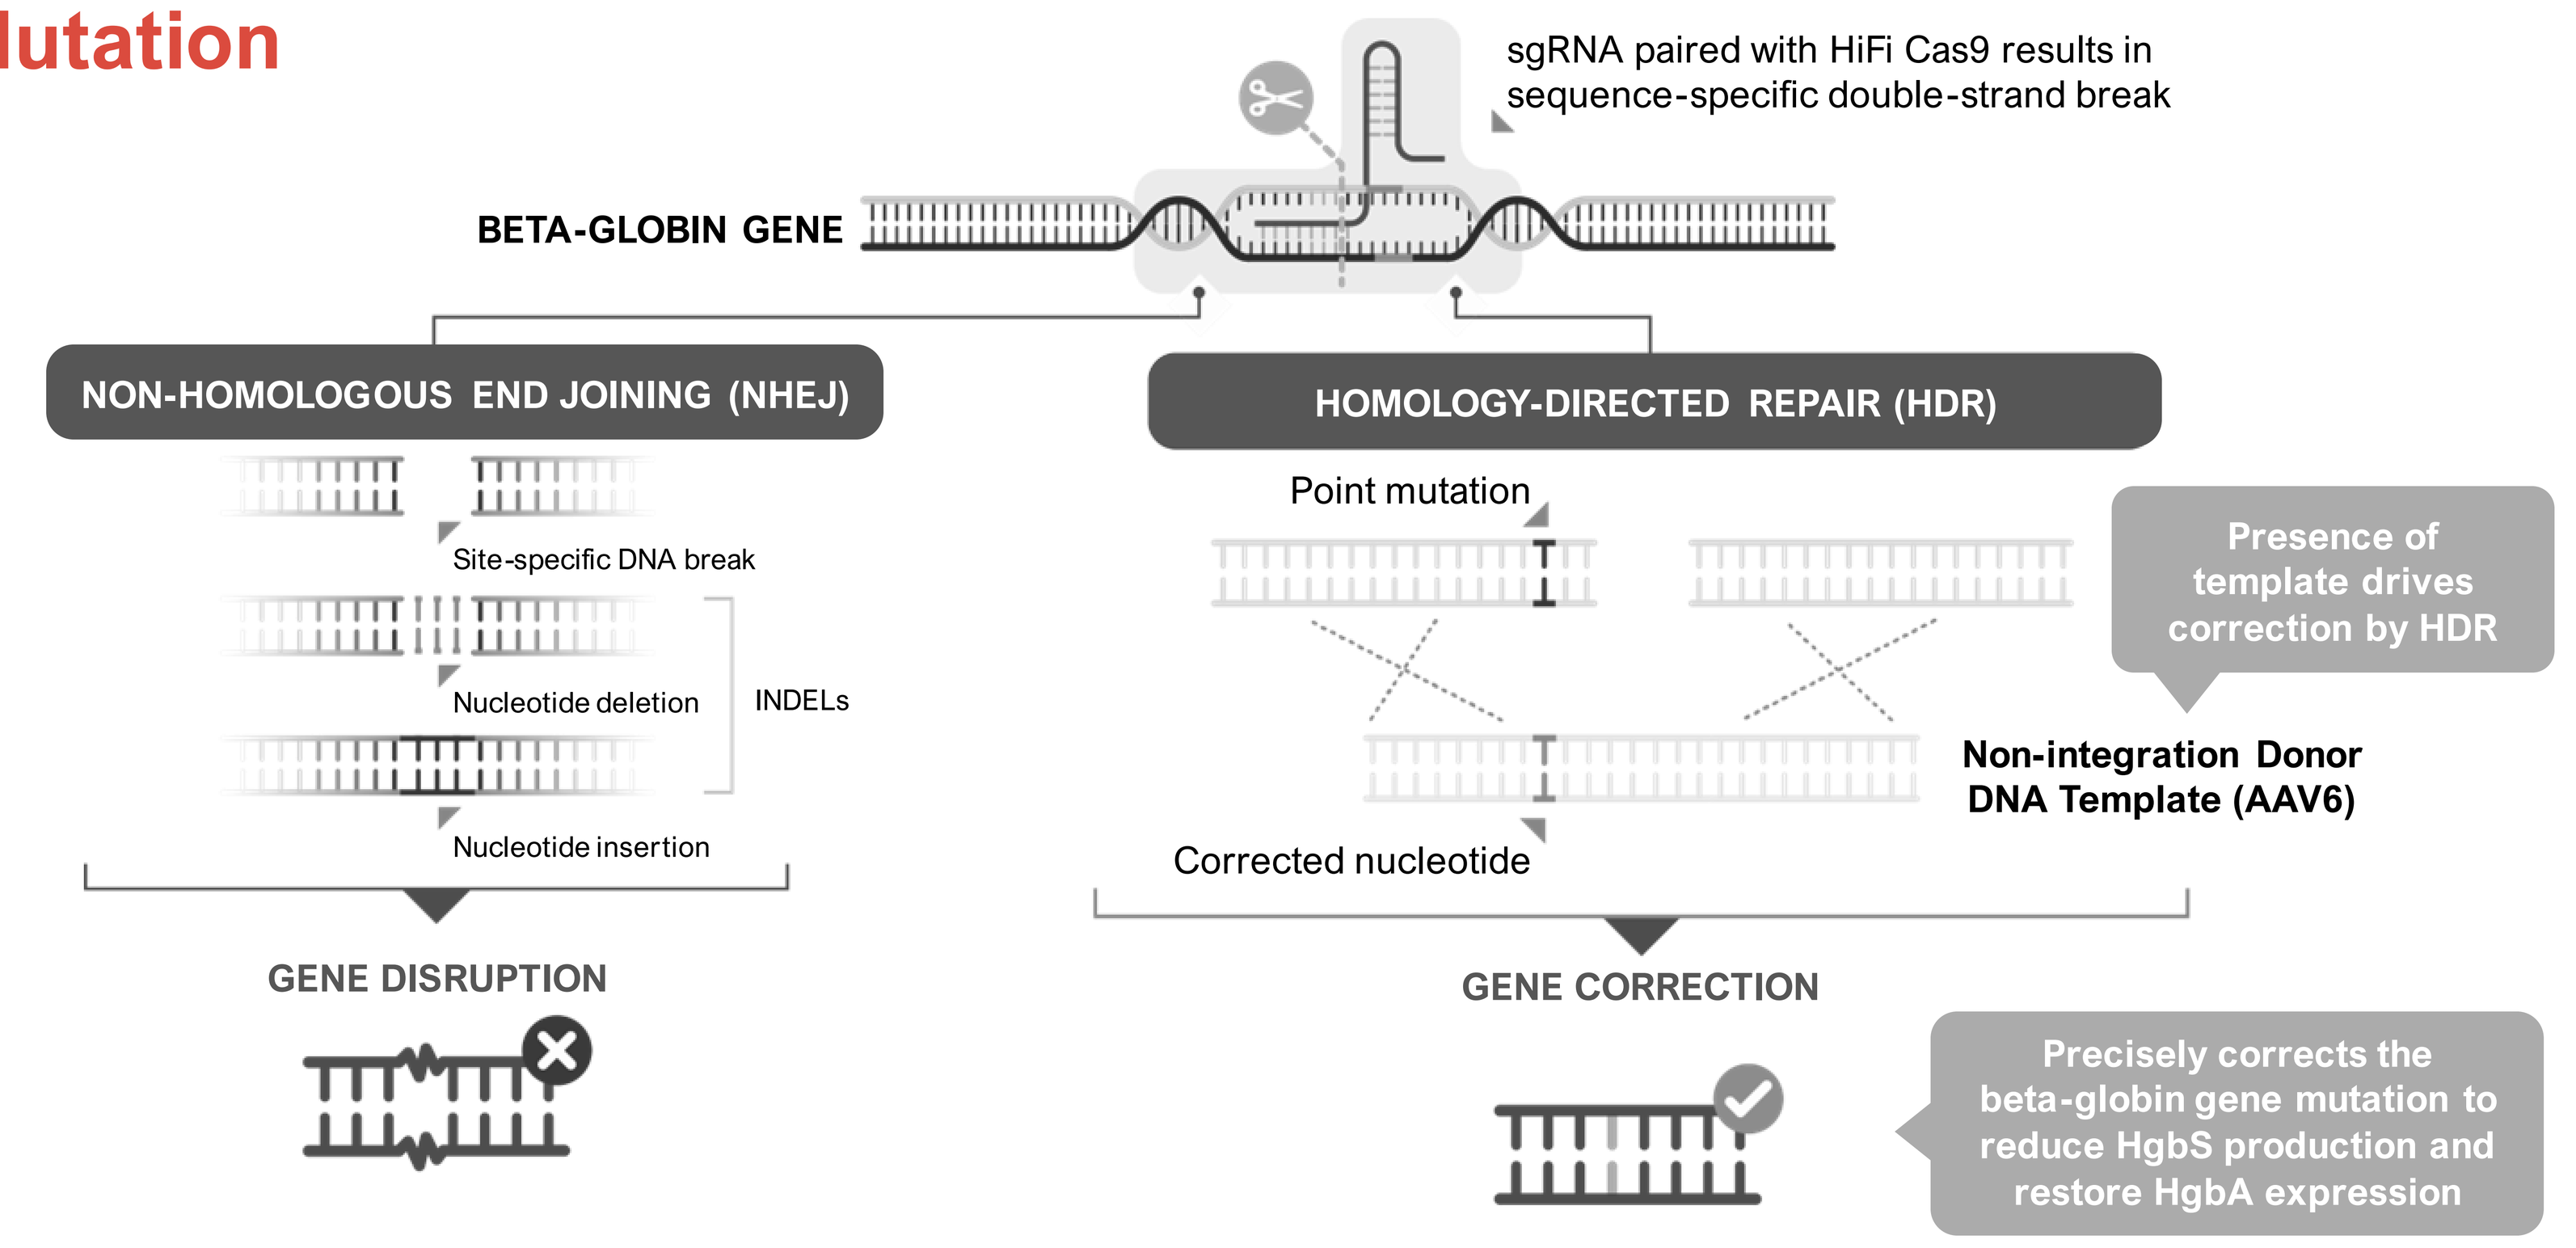

sgRNA paired with HiFi Cas9 results in sequence-specific double-strand break
BETA-GLOBIN GENE
NON-HOMOLOGOUS END JOINING (NHEJ)
HOMOLOGY-DIRECTED REPAIR (HDR)
Site-specific DNA break
Nucleotide deletion
INDELs
Nucleotide insertion
GENE DISRUPTION
Point mutation
Presence of template drives correction by HDR
Non-integration Donor DNA Template (AAV6)
Corrected nucleotide
GENE CORRECTION
Precisely corrects the beta-globin gene mutation to reduce HgbS production and restore HgbA expression

AAV6, adeno-associated virus type 6; Cas9, CRISPR-associated protein 9; CRISPR, clustered regularly interspaced short palindromic repeats; HgbA, adult hemoglobin; HgbS, sickle hemoglobin; HDR, homology-directed repair; HiFi, high fidelity; INDEL, insertion and/or deletion; NHEJ, non-homologous end joining; SCD, sickle cell disease; sgRNA, single-strand guide RNA.

Clinical Protocol

Schema for Screening, Mobilization, Apheresis, and Transplant

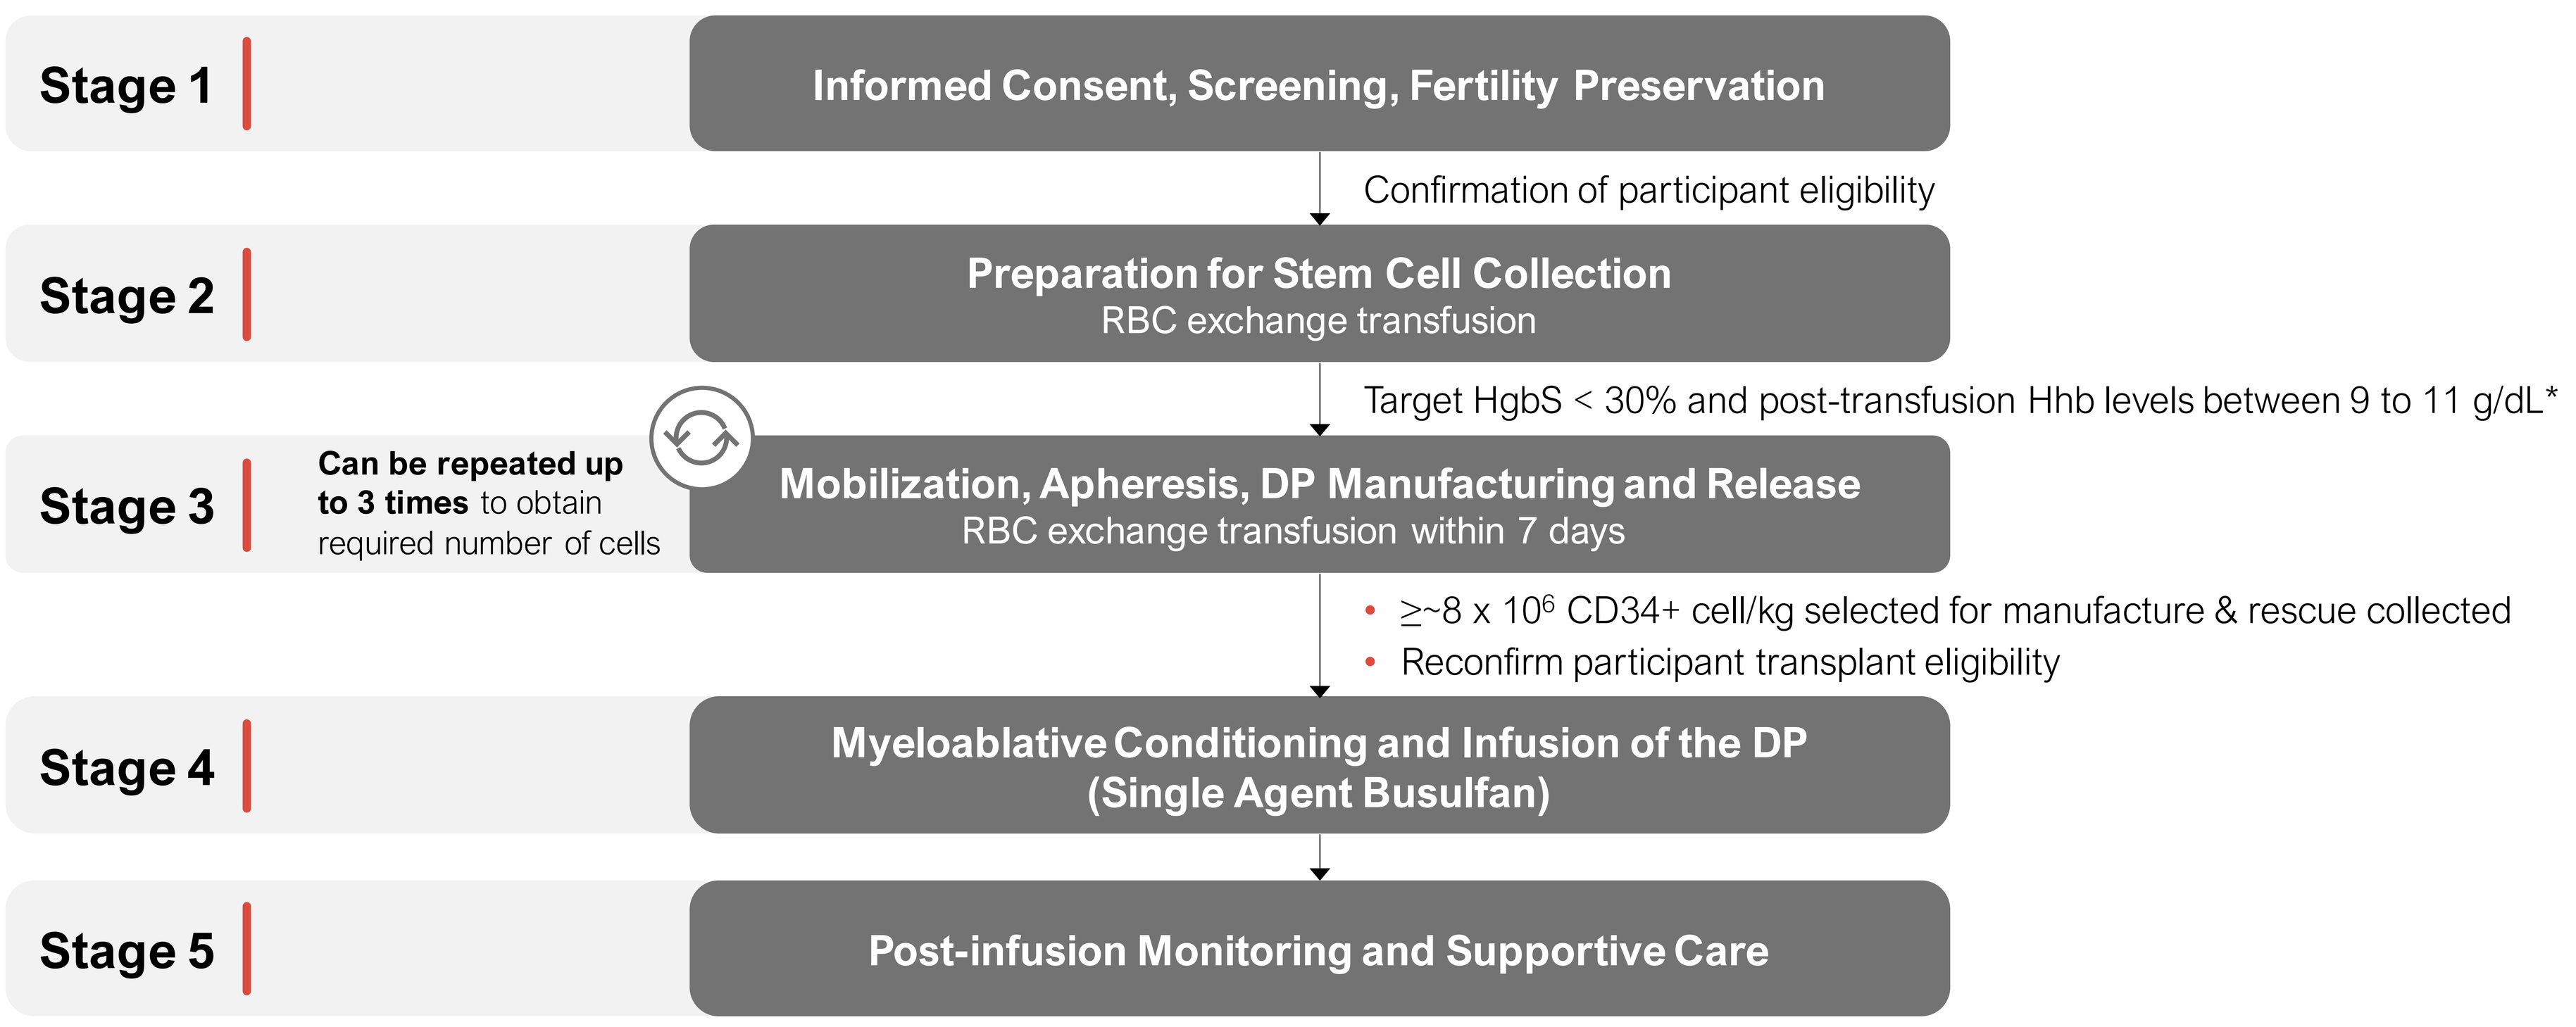

Stage 1
Informed Consent, Screening, Fertility Preservation
Confirmation of participant eligibility
Stage 2
Preparation for Stem Cell Collection
RBC exchange transfusion
Target HgbS < 30% and post-transfusion Hhb levels between 9 to 11 g/dL*
Stage 3
Can be repeated up to 3 times to obtain required number of cells
Mobilization, Apheresis, DP Manufacturing and Release
RBC exchange transfusion within 7 days
≥~8 x 10⁶ CD34+ cell/kg selected for manufacture & rescue collected
Reconfirm participant transplant eligibility
Stage 4
Myeloablative Conditioning and Infusion of the DP
(Single Agent Busulfan)
Stage 5
Post-infusion Monitoring and Supportive Care

***Hgb level for last exchange transfusion before each mobilization cycle should target between 8 to 10 g/dL.**

Abbreviations: CD, cluster of differentiation; DP, Drug product; Hgb, hemoglobin; HgbS, hemoglobin S; LTFU, long-term follow-up; RBC, red blood cell, SoA, schedule of assessment.

Participation in a LTFU study (for a total follow-up of 15 years after DP infusion) after will be offered to participants after completion of this protocol

Phase 1 Trial Design

Evaluate the safety of treatment with KMAU-001 in participants with severe SCD

Primary Objectives	Secondary Objectives	Exploratory Objectives
Evaluate the safety of treatment with KMAU-001 in participants with severe SCD	**Evaluate the efficacy and pharmacodynamics of treatment with KMAU-001 in participants with severe SCD**	**Evaluate PROs, erythrocyte function, characterization of gene correction rates, and change from baseline in select SCD characteristics and organ function**
• Proportion of patients who reach neutrophil engraftment within 42 days post infusion • Treatment-related mortality • Overall survival • Frequency and severity of AEs including clinically relevant lab abnormalities	• Time to neutrophil, platelet engraftment • Assessment of the following over time post DP Infusion: – % of adult Hgb and HbS of total Hgb – Proportion of participants achieving HbS < 50% for at least 3 months – Peripheral myeloid cells gene correction levels – Globin gene expression compared to baseline – Proportion of complete resolution of VOE and incidence rate of VOE – Changes in pRBC transfusion needs (volume and frequency)	• Cerebral hemodynamics and oxygen delivery (by MRA/MRI) • Improvements in SCD-related events and changes in organ function(e.g. heart, brain, liver) • Measurements of RBC health and function • Correlation of KMAU-001 DP characteristics with clinical outcomes • Changes in VOC and ACS post infusion compared to medical history • Measurement of INDELs, off-target editing, and gene correction levels in peripheral immune cells

Patient 1: Clinical History and Clinical Course

23-year-old African American Female with Hemoglobin S/S and heterozygous 1 gene deletion (3.7kb deletion) of alpha globin genes.

Prior to study enrollment, patient experienced ~5 vaso-occlusive episodes (VOEs) per year requiring hospitalization for treatment.

She had no history of stroke or other major complications of sickle cell disease.

Summary of Drug Product Infusion:

- Busulfan CSS ~770
- Total Cell Dose Manufactured: 8.75 x 10^6 CD34/kg
- Viable Cell Dose Infused: 3.5 x 10^6 CD34/kg

Bone Marrow Examinations on D+ 91, 135, 184, 364

- On D+91 the marrow was hypocellular but improved by D+135 showing erythroid hyperplasia with no overt dysplasia or evidence of malignant progression

TABLE 1: Allele Editing Frequencies at On and Off-Target Sites

Day After Infusion	Sample	INDELs (%)	Unedited (%)	Corrected (%)	INDELs OT1 (%)
Pre-Infusion	Drug Product (nula-cel)	21.4	43	33.1	0.88
Day 37	PB CD15+	66.92	18.93	9.31	2.3
Day 49	PB CD15+	69.93	20.43	6.68	2.47
Day 63	PB CD15+	70.66	20.78	5.89	2.3
Day 91	PB CD15+	70.61	20.72	5.93	2.32
	BMMC	69.36	25.25	3.39	2.34
	CD34+	71.31	21.15	5.17	Not Done
Day 126	PB CD15+	68.13	23.38	6.05	2.46
Day 135	BMMC	66.54	28.87	3.32	1.69
	CD34+	74.31	18.80	5.27	Not Done
Day 182	PB CD15+	70.2	21.3	5.8	2.43
Day 184	BMMC	73.6	21.4	2.3	2.13
Day 364	PB CD15+	92.6	5.4	1.3	0.62

Abbreviations: PB CD15+=Peripheral Blood CD15+ Purified Cells; BMMC=Bone marrow mononuclear cells; CD34+=Bone Marrow CD34+ Purified Cells.

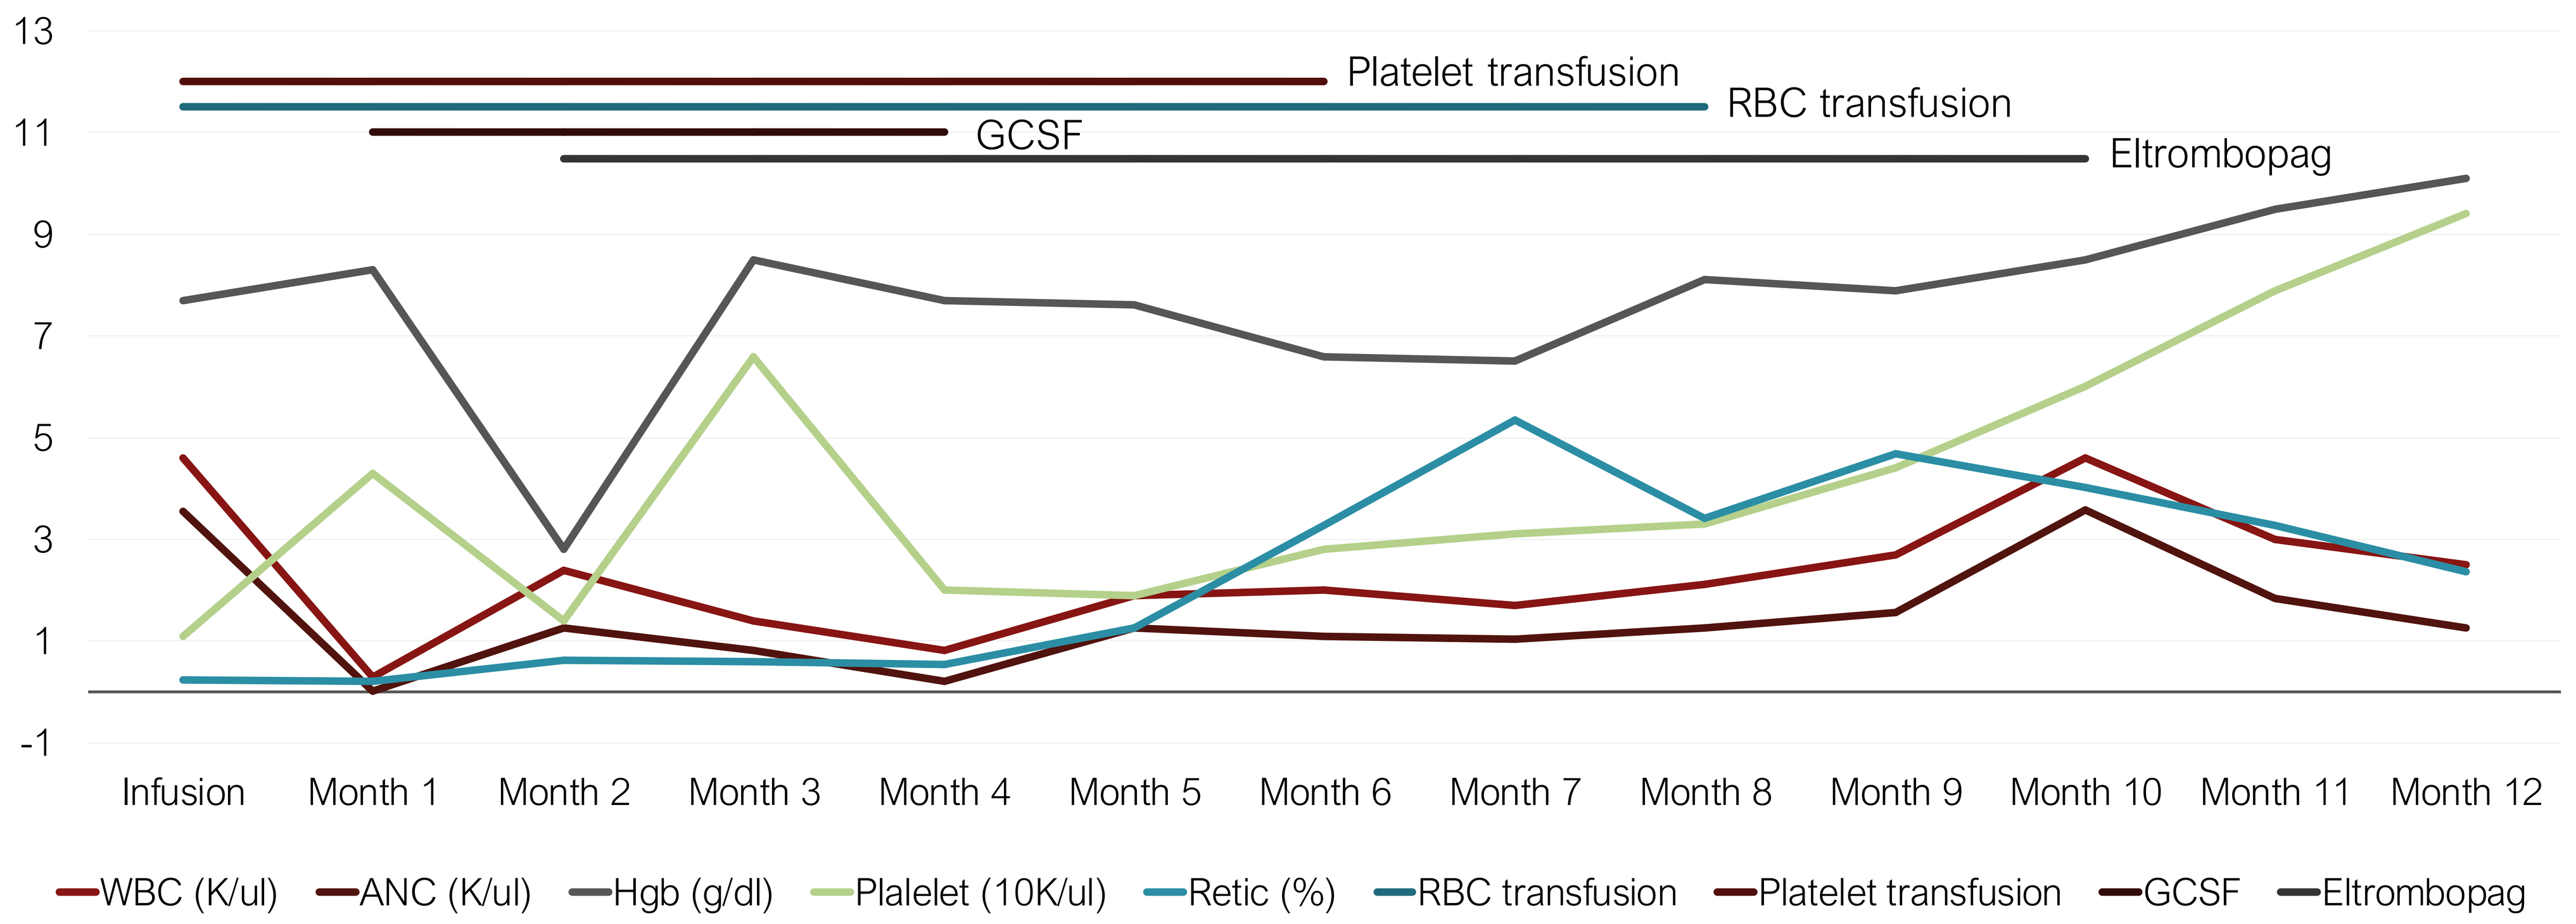

13
11
9
7
5
3
1
-1
Platelet transfusion
RBC transfusion
GCSF
Eltrombopag
Infusion
Month 1
Month 2
Month 3
Month 4
Month 5
Month 6
Month 7
Month 8
Month 9
Month 10
Month 11
Month 12
WBC (K/ul)
ANC (K/ul)
Hgb (g/dl)
Plalelet (10K/ul)
Retic (%)
RBC transfusion
Platelet transfusion
GCSF
Eltrombopag

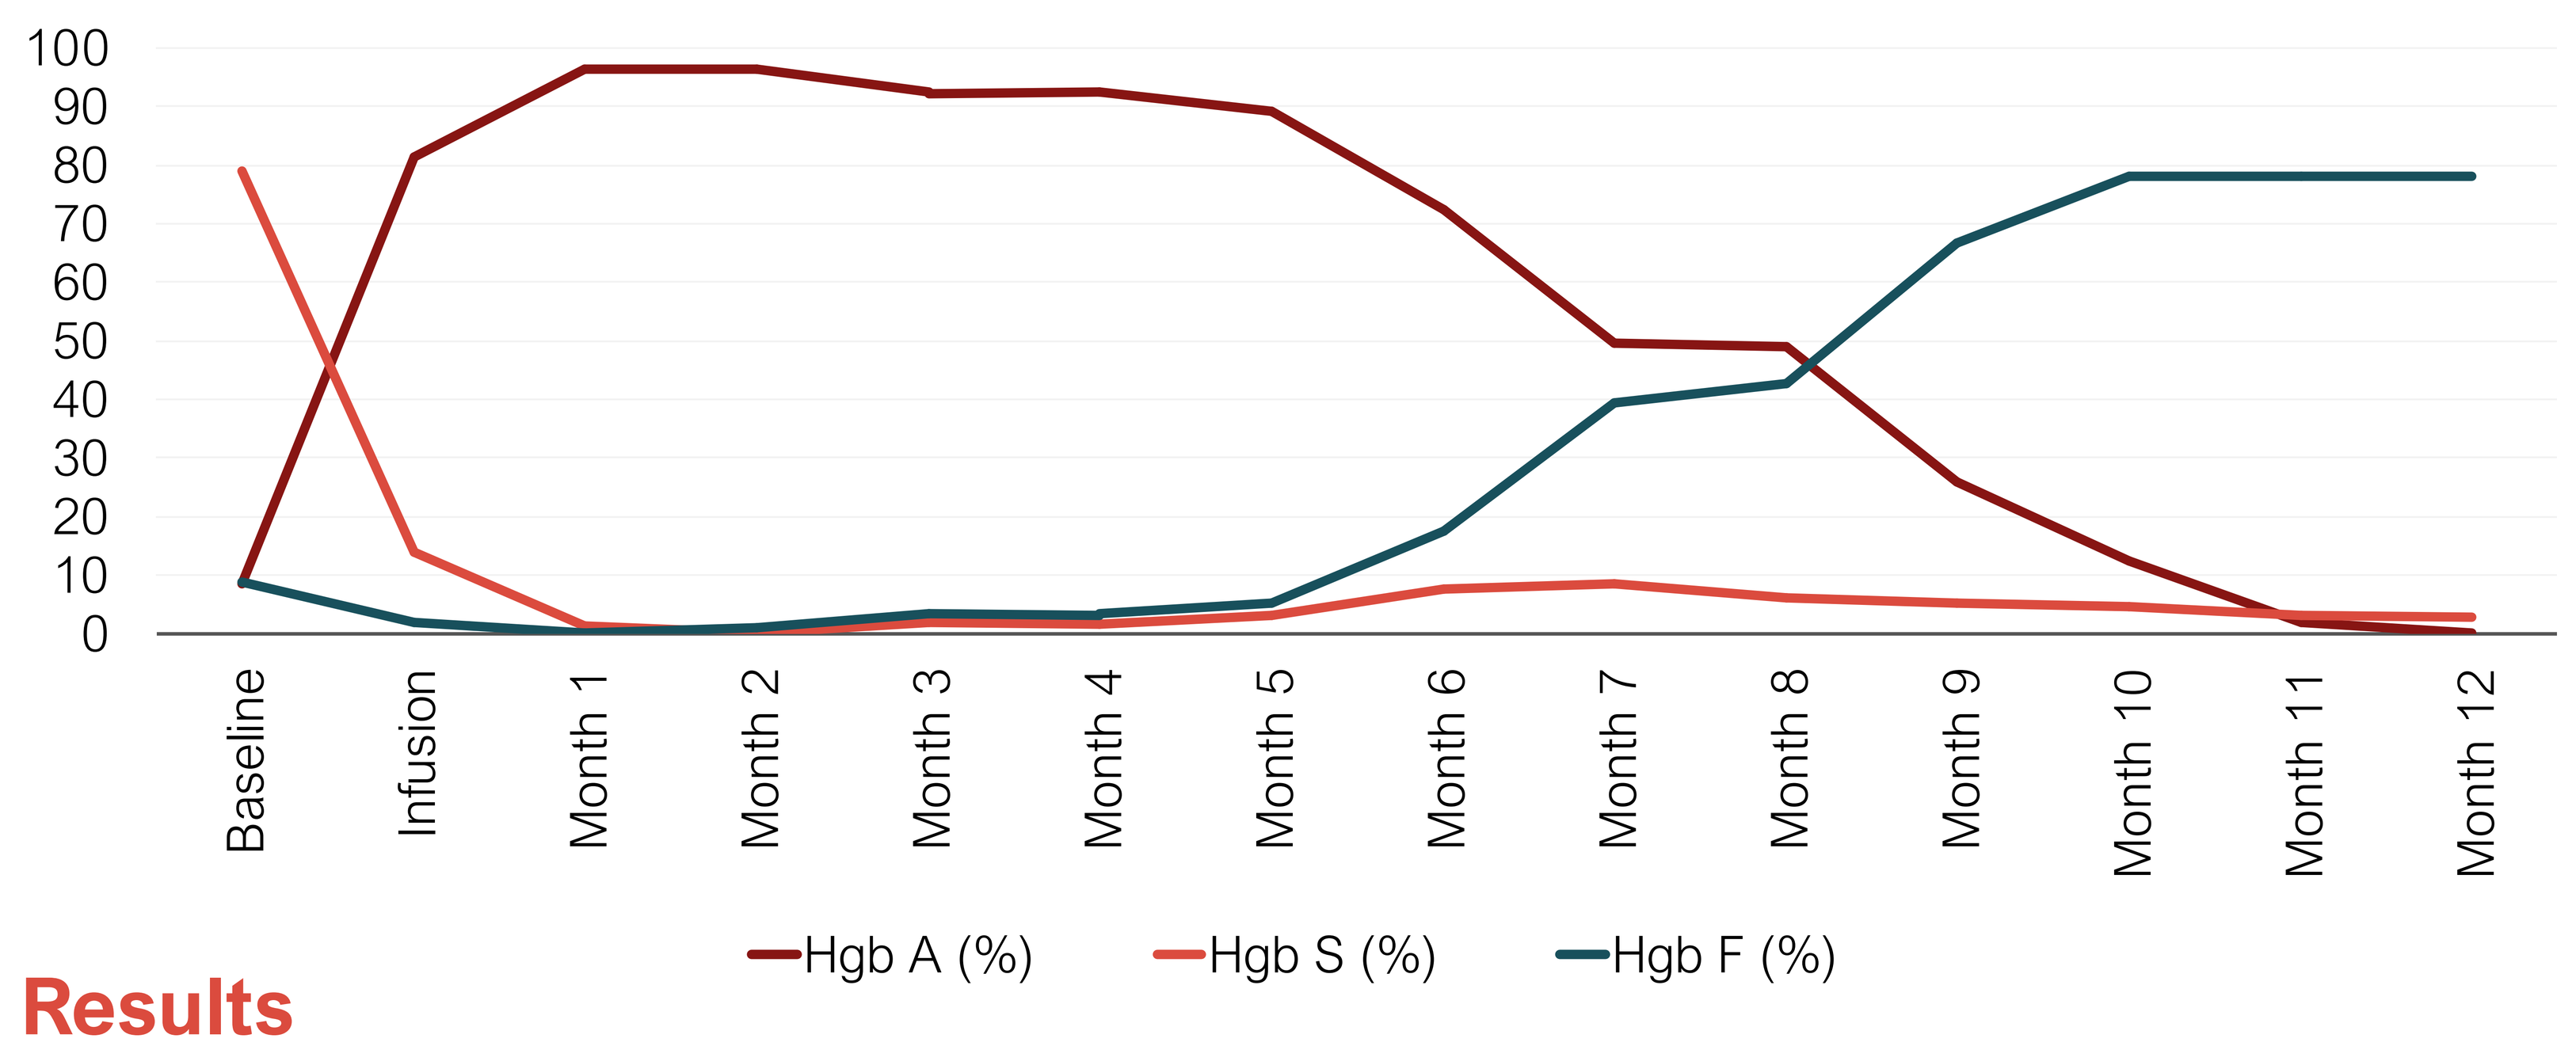

100
90
80
70
60
50
40
30
20
10
0
Baseline
Infusion
Month 1
Month 2
Month 3
Month 4
Month 5
Month 6
Month 7
Month 8
Month 9
Month 10
Month 11
Month 12
Hgb A (%)
Hgb S (%)
Hgb F (%)

Results

Patient had no VOEs, greatly improved QOL and reduced chronic pain post DP infusion

Transfusion iron overload improving on deferasirox.

The patient has remained RBC transfusion free since month 8 and narcotics for SCD related chronic pain has been weaned.

12-month Hemoglobin Distribution:

HgbA=1.9%, HgbS=3.1%, HgbF=>78%

A BCOR splice site variant (c.4326+1G>A) was detected by hybrid capture next generation sequencing from blood/marrow samples suggesting clonal hematopoiesis without marrow dysplasia or cytogenetic abnormality at months 6 (VAF 9%) and month 12 (VAF 23%).

- BCOR variants are found in aplastic anemia (where they can rise and fall[1]) and following autologous transplant using unmanipulated cells[2]
- No evidence that genome editing caused the BCOR variant and the variant site is not an off-target site for this gRNA
- BCOR VAF as well as hematopoiesis and marrow morphology will be followed regularly

Bone Marrow Biopsy Morphology: 10X magnification demonstrating excellent overall cellularity, no fibrosis and 40X magnification demonstrating no dysplasia.

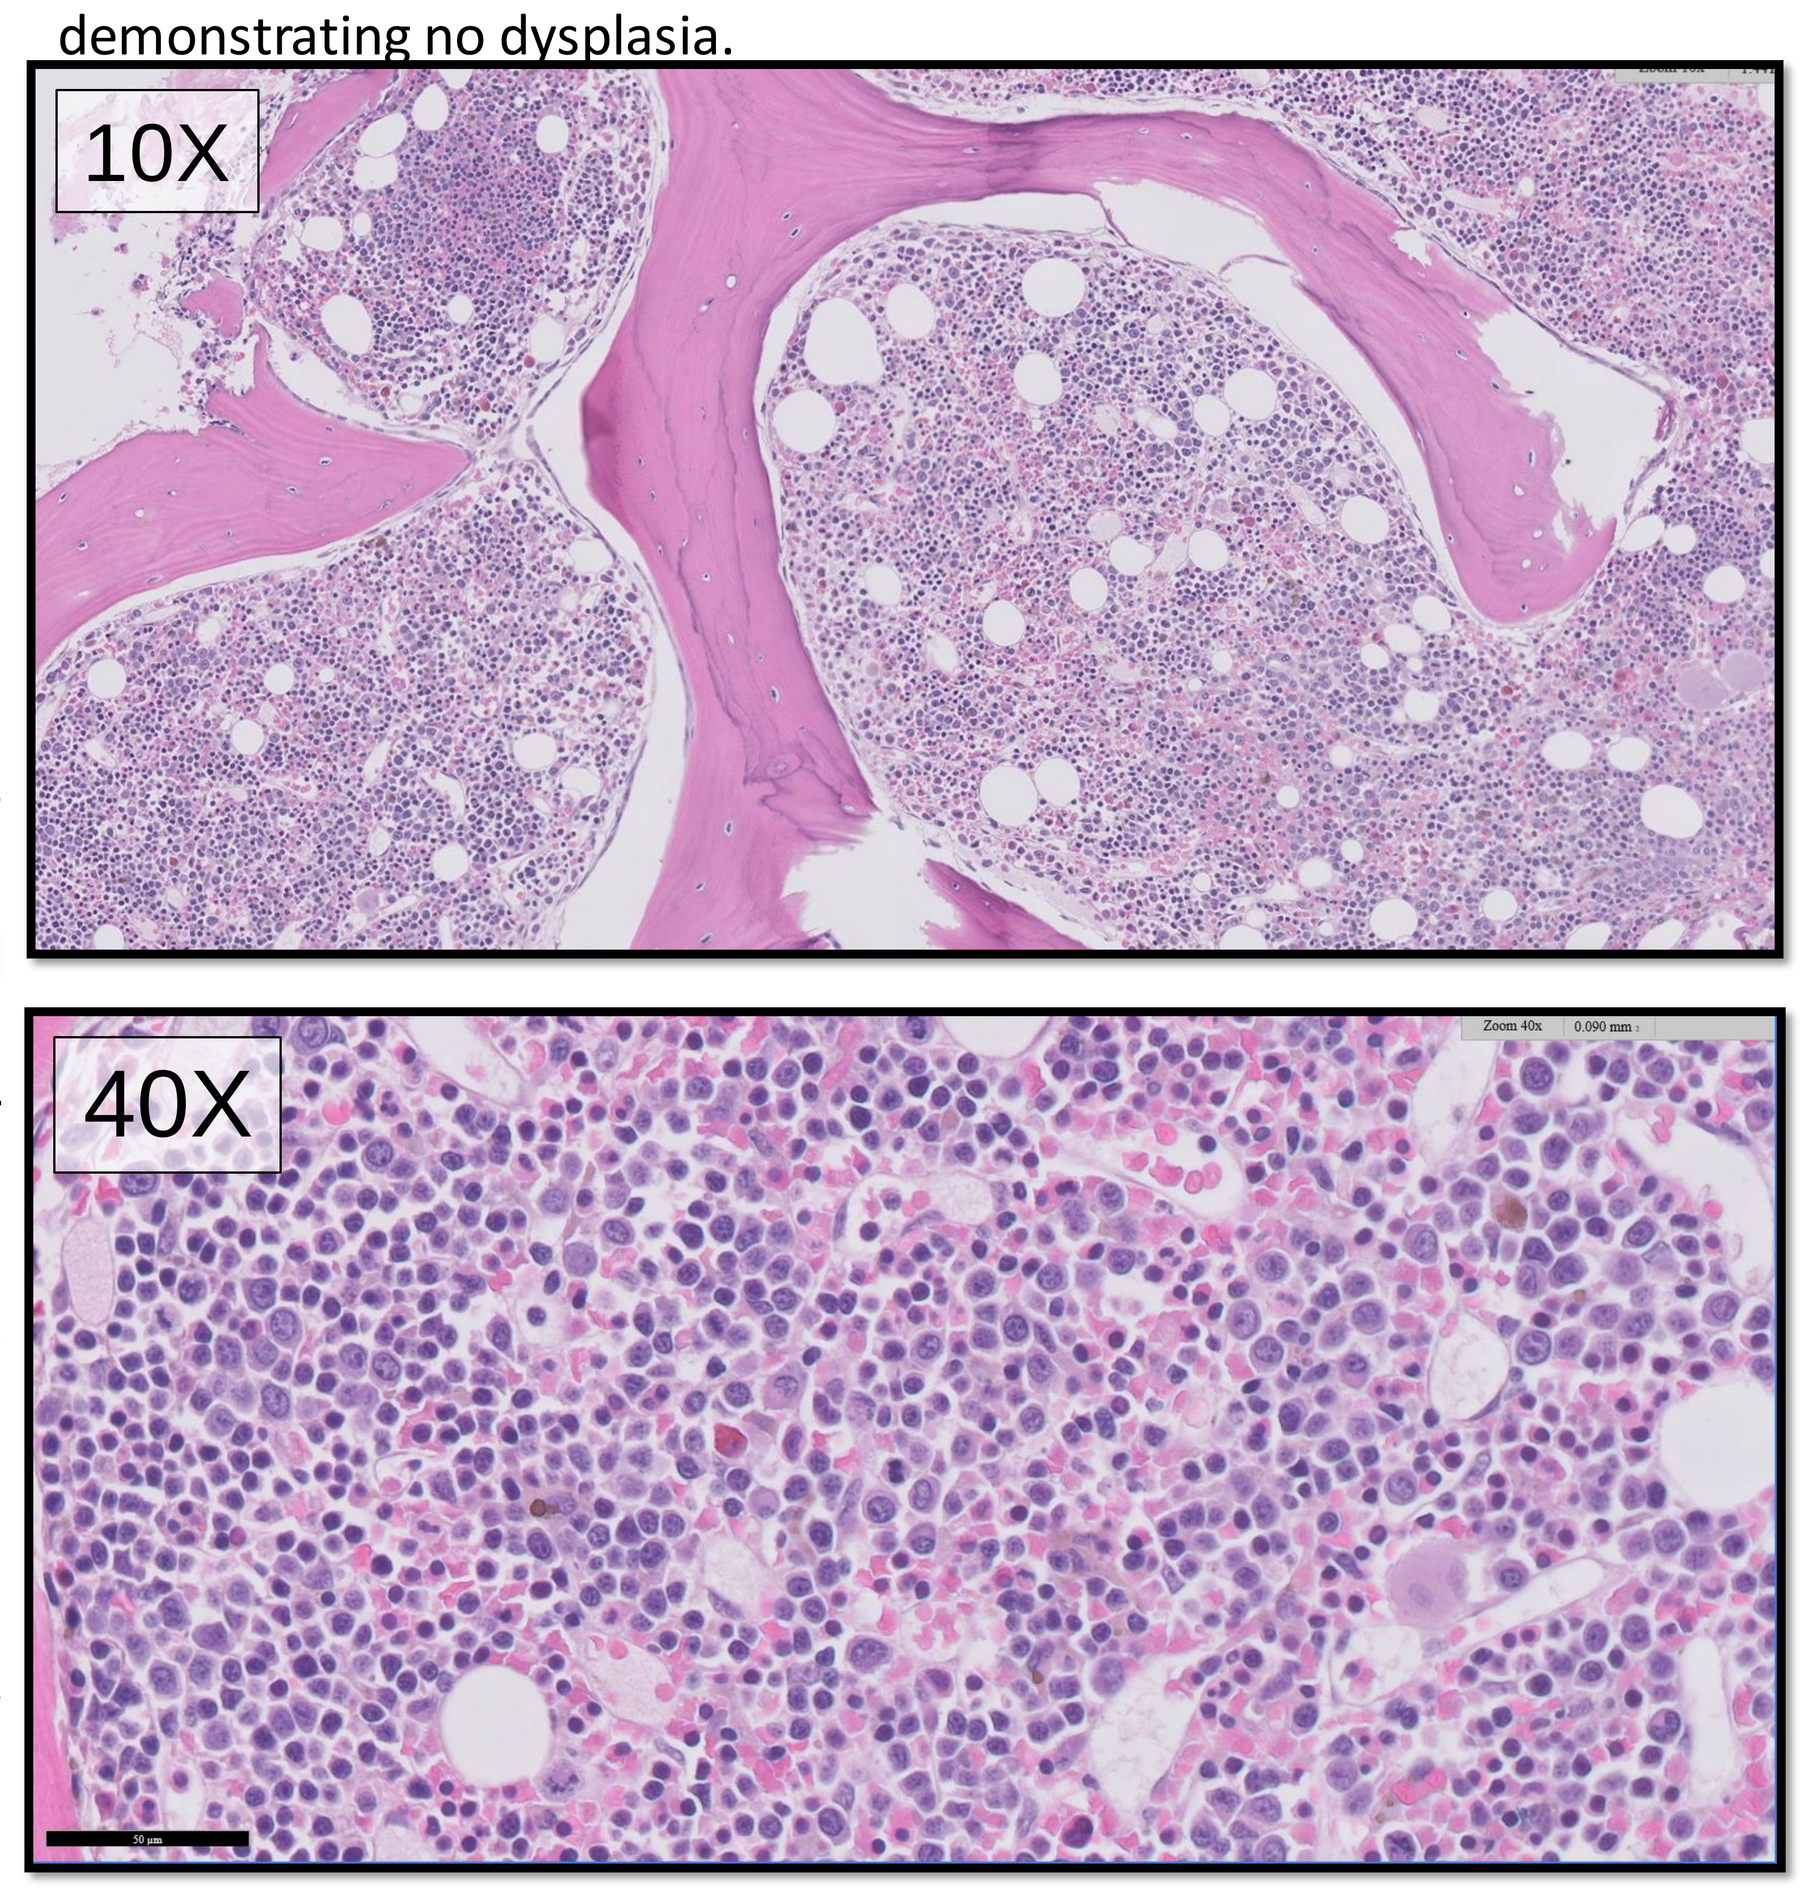

10X
40X

Discussion

Patient shows a unique hemoglobin profile compared to all other gene edited/gene therapies for sickle cell disease with HgbS percentage <5% and HgbF >78% without transfusion.

The DP had an unexpected mechanism of therapeutic benefit. The mechanism of HgbF upregulation/production is currently under investigation.

HBB genotype of BCOR variant containing clone is under investigation.

Clone does not contain an INDEL at OT1 by inference (OT1 INDEL frequency fell as clone expanded), experimental confirmation pending.

Manufacturing improvements, to reduce drive induced by stress hematopoiesis, include the addition of an HDR booster, refined culturing conditions, and shortened duration of manufacturing have resulted in improved cell yields and cell quality and will be used to manufacture the DP for the next patients.

Long term follow up protocol submitted to FDA.

References

1. N Engl J Med 2015; 373:35-47 2. Cell Reports 2019; 27:2022-2028.